TTX-080, A First-in-Class HLA-G Specific Antagonist, Increases Distinct Innate and Adaptive Immune Cells in the Tumor Microenvironment and Periphery

Courtney Beers, PhD[1], Megan Welch[1], Stephanie Adams[1], Anthony Desbien, PhD[1], and Douglas Hodges[1]

[1]Tizona Therapeutics, South San Francisco, CA, United States

Abstract 570

Background

- Human leukocyte antigen G (HLA-G), a non-classical MHC class I molecule expressed on solid tumors, is known to drive immune suppression, promote cancer cell immune escape, and lead to tumor development and growth. HLA-G mediates suppression through ILT2 and ILT4 on adaptive (ILT2) and innate (ILT2 & ILT4) immune cell subpopulations. Blocking HLA-G has the potential to reverse immune tolerance and activate anti-tumor immune responses
- TTX-080 is a novel, first-in-class, fully human monoclonal antagonistic antibody designed to specifically bind to HLA-G and block interactions with both ILT2 and ILT4
- TTX-080 is currently being evaluated in a randomized Phase 1b study in combination with cetuximab + FOLFIRI in patients with metastatic colorectal cancer, based on anti-tumor activity observed in a single-arm study of TTX-080. (Ulahannan, et. al., ASCO 2024)
- Here, we present translational data from the single-arm Phase 1b clinical trial of TTX-080 as monotherapy and in combination with pembrolizumab, cetuximab (anti-EGFR IgG1:FcγR ADCC & ADCP competent), and cetuximab + FOLFIRI in patients with metastatic or advanced cancer
- TTX080 demonstrated statistically significant on-mechanism activation of innate and adaptive immune cells in the tumor microenvironment and in the periphery validating the functional impact of HLA-G blockade on immune cells in a clinical setting

Figure 1. HLA-G Suppresses both Adaptive & Innate Immune Activity

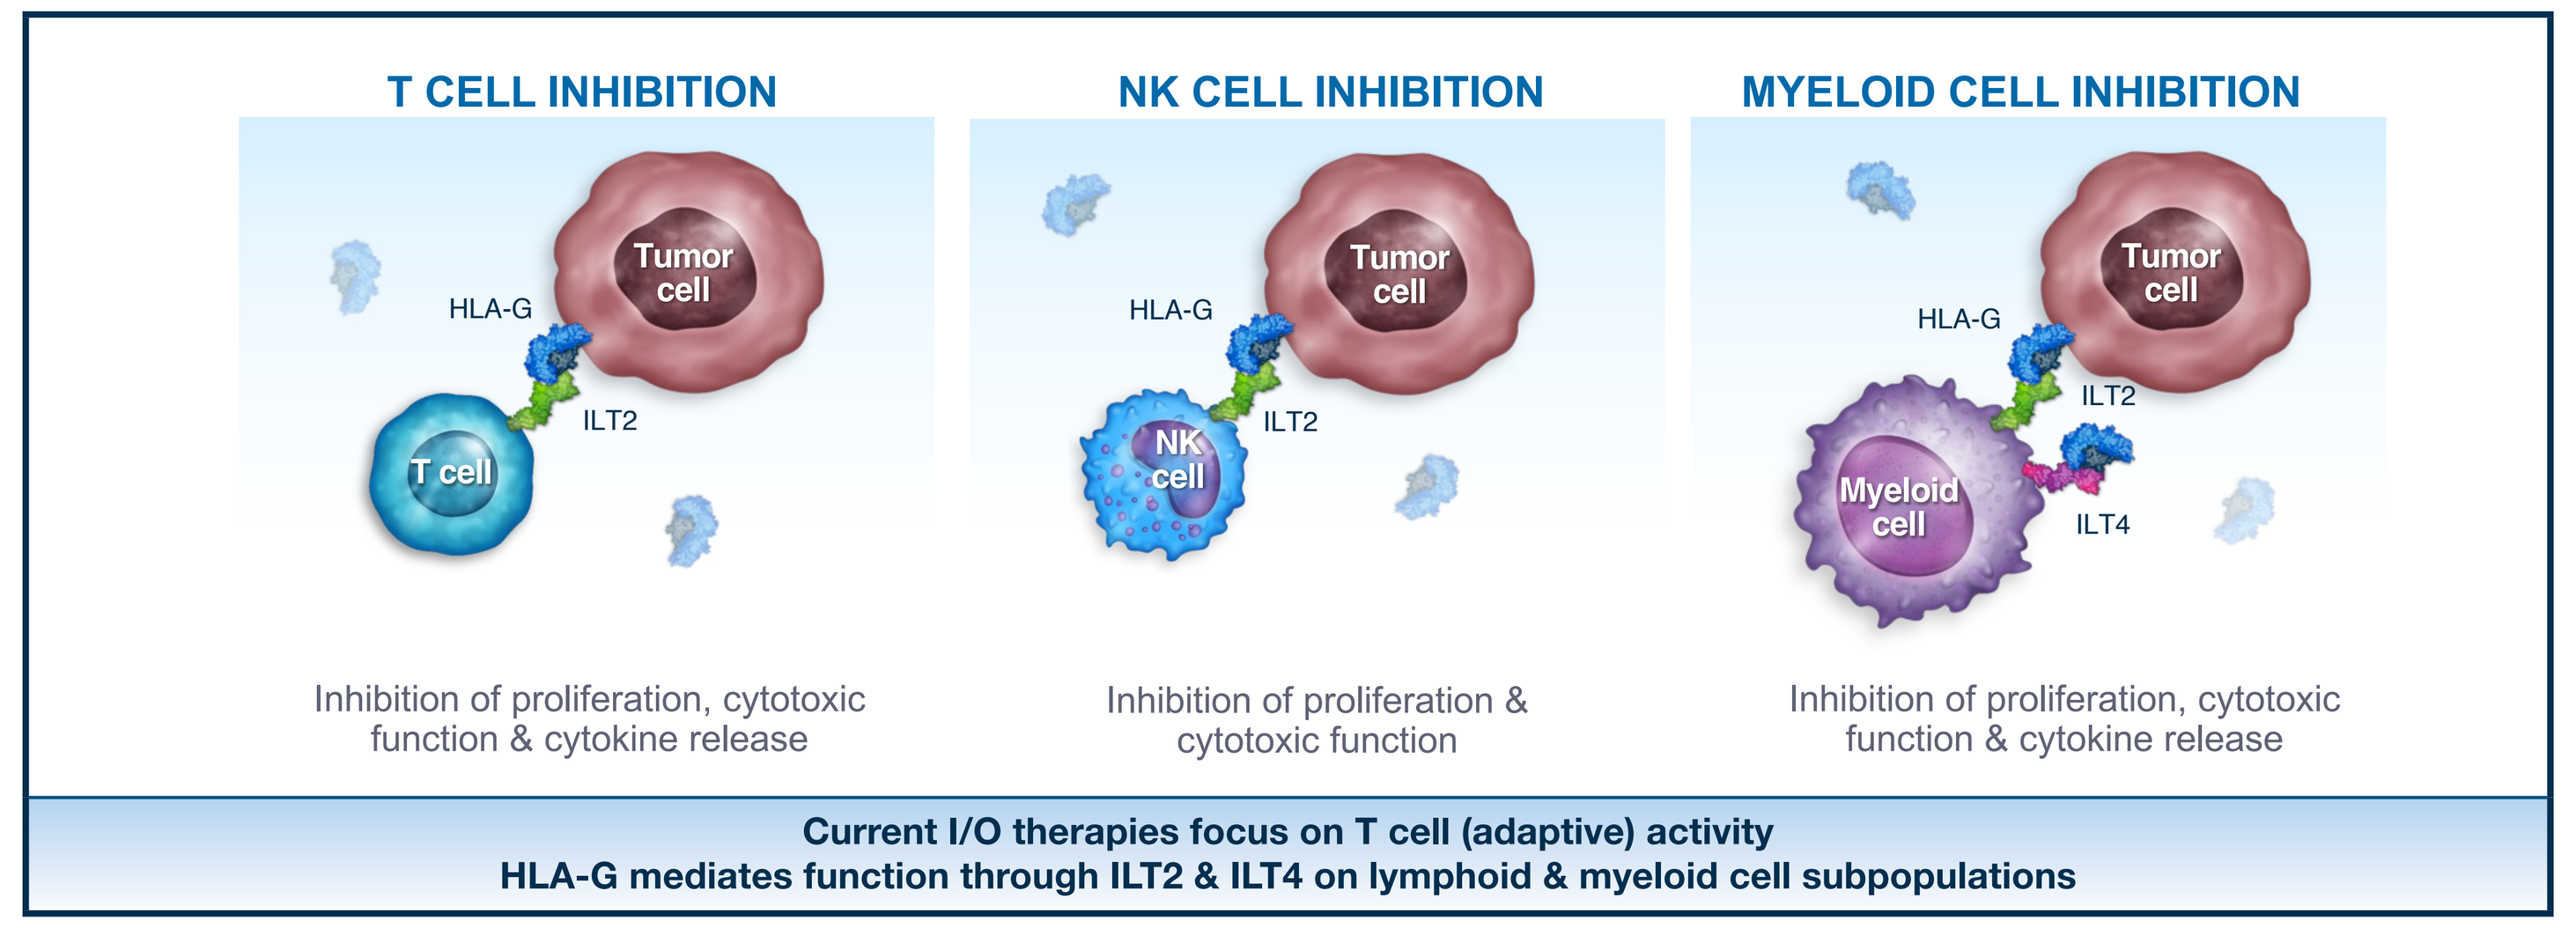
T CELL INHIBITION
NK CELL INHIBITION
MYELOID CELL INHIBITION
Tumor cell
HLA-G
ILT2
ILT4
T cell
NK cell
Myeloid cell
Inhibition of proliferation, cytotoxic function & cytokine release
Inhibition of proliferation & cytotoxic function
Inhibition of proliferation, cytotoxic function & cytokine release
Current I/O therapies focus on T cell (adaptive) activity
HLA-G mediates function through ILT2 & ILT4 on lymphoid & myeloid cell subpopulations

Figure 2. TTX-080 has the Potential to Enhance Anti-Tumor Responses

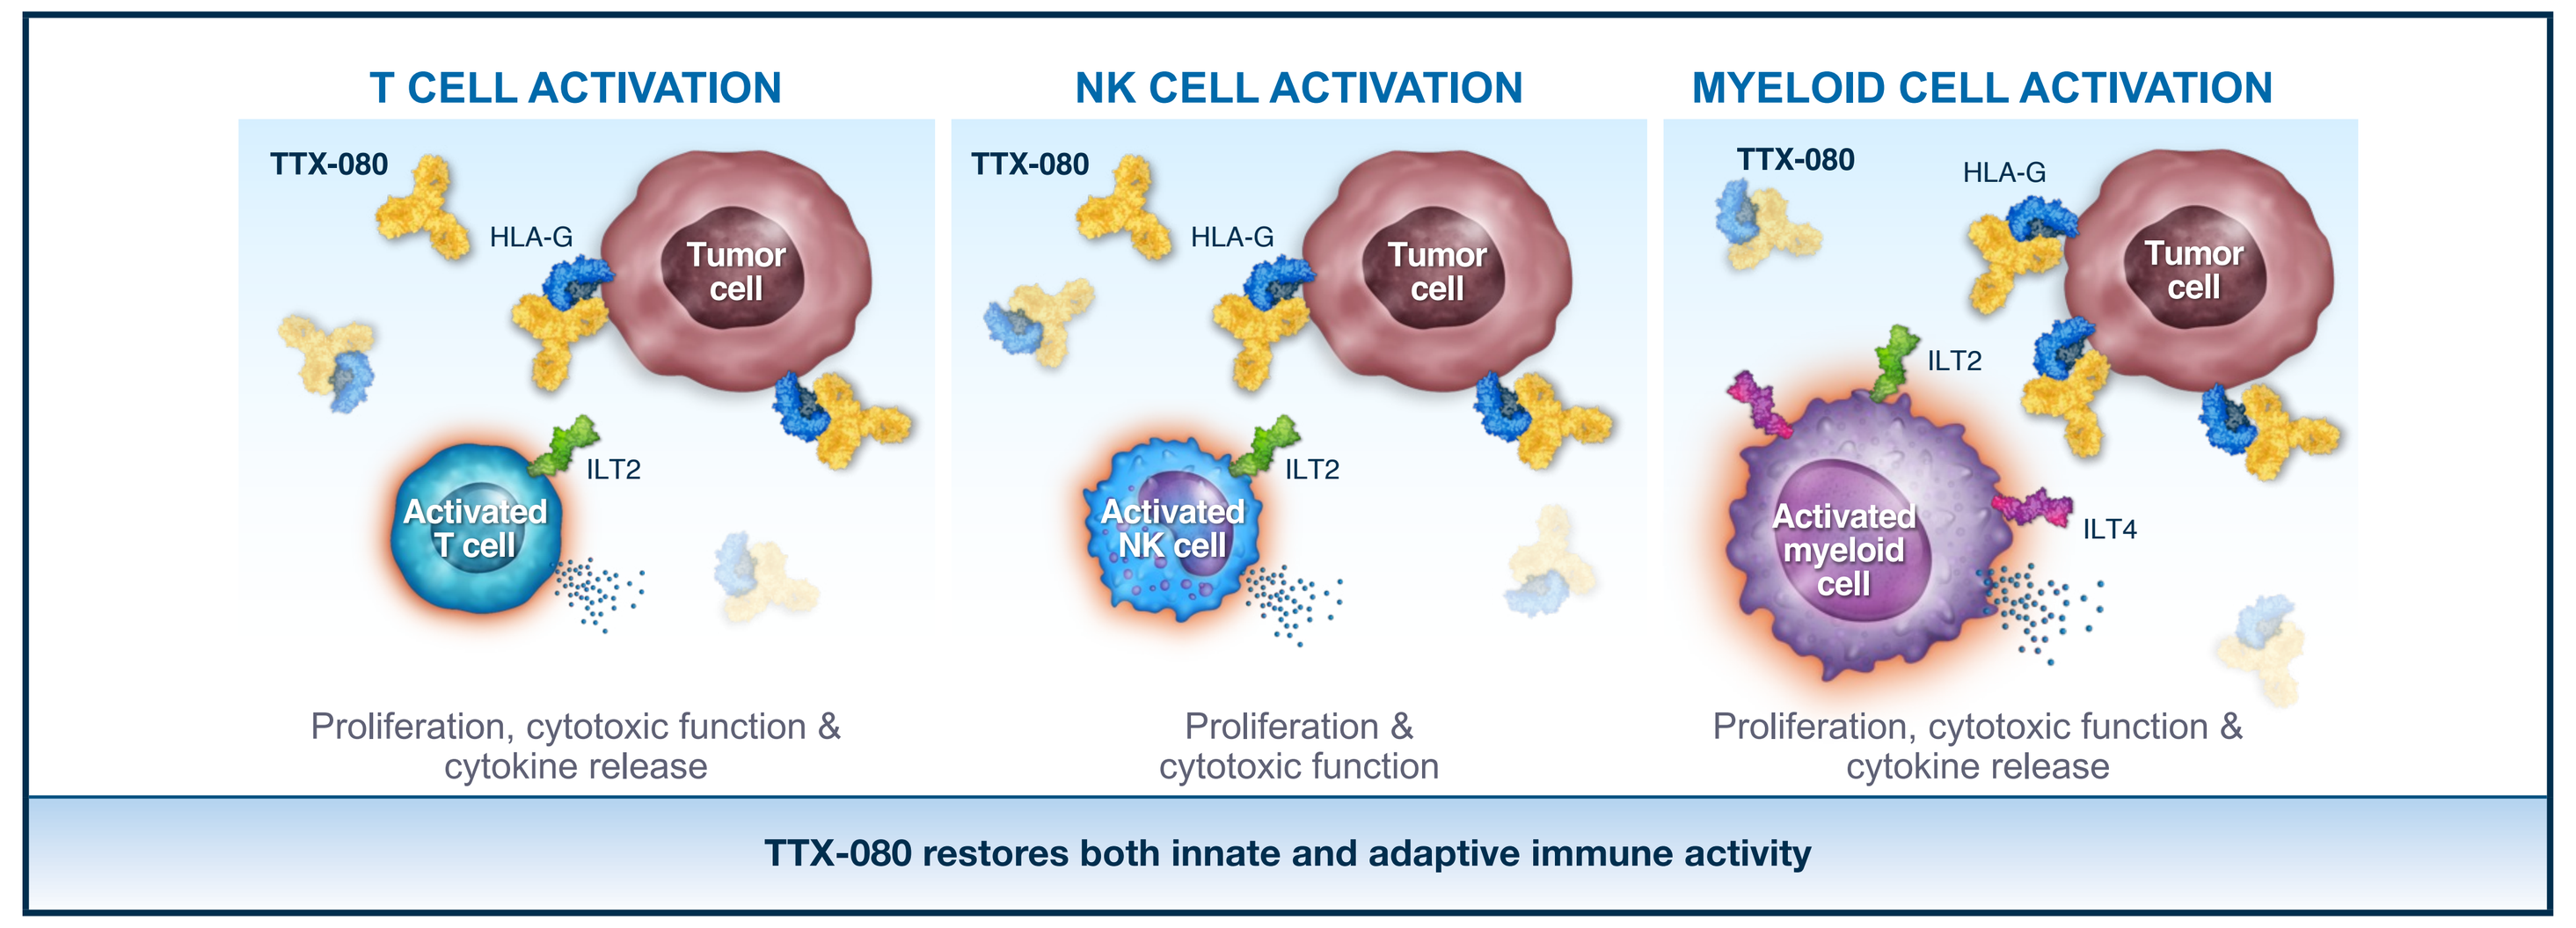
T CELL ACTIVATION
NK CELL ACTIVATION
MYELOID CELL ACTIVATION
TTX-080
HLA-G
Tumor cell
ILT2
ILT4
Activated T cell
Activated NK cell
Activated myeloid cell
Proliferation, cytotoxic function & cytokine release
Proliferation & cytotoxic function
Proliferation, cytotoxic function & cytokine release
TTX-080 restores both innate and adaptive immune activity

Figure 3. TTX-080's Path to Clinical Proof of Concept

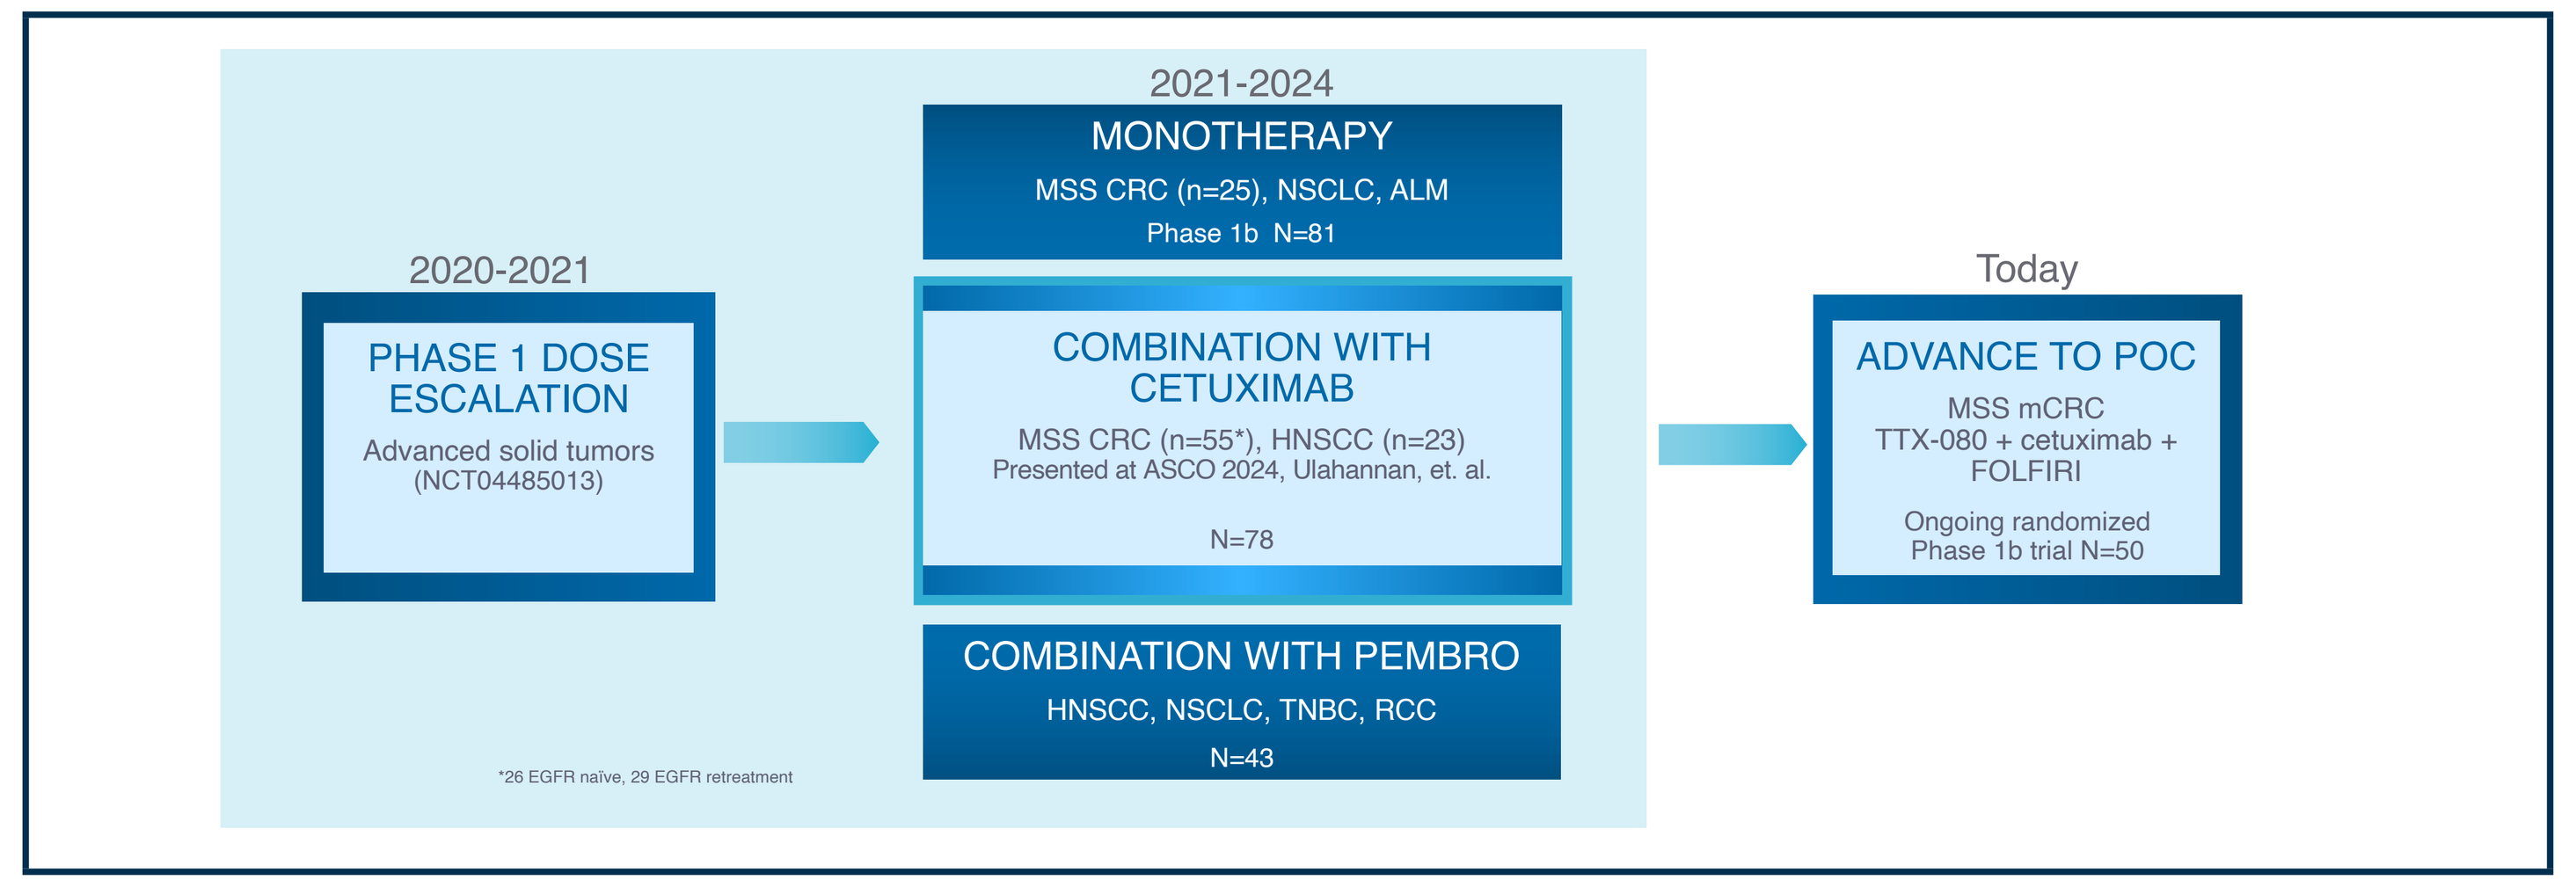
2020-2021
PHASE 1 DOSE ESCALATION
Advanced solid tumors (NCT04485013)
2021-2024
MONOTHERAPY
MSS CRC (n=25), NSCLC, ALM
Phase 1b N=81
COMBINATION WITH CETUXIMAB
MSS CRC (n=55*), HNSCC (n=23)
Presented at ASCO 2024, Ulahannan, et. al.
N=78
COMBINATION WITH PEMBRO
HNSCC, NSCLC, TNBC, RCC
N=43
*26 EGFR naïve, 29 EGFR retreatment
Today
ADVANCE TO POC
MSS mCRC
TTX-080 + cetuximab + FOLFIRI
Ongoing randomized Phase 1b trial N=50

Results

Figure 4. TTX-080 Exhibits Well-Behaved PK in Phase 1 Patients and Supports Q3W Dosing

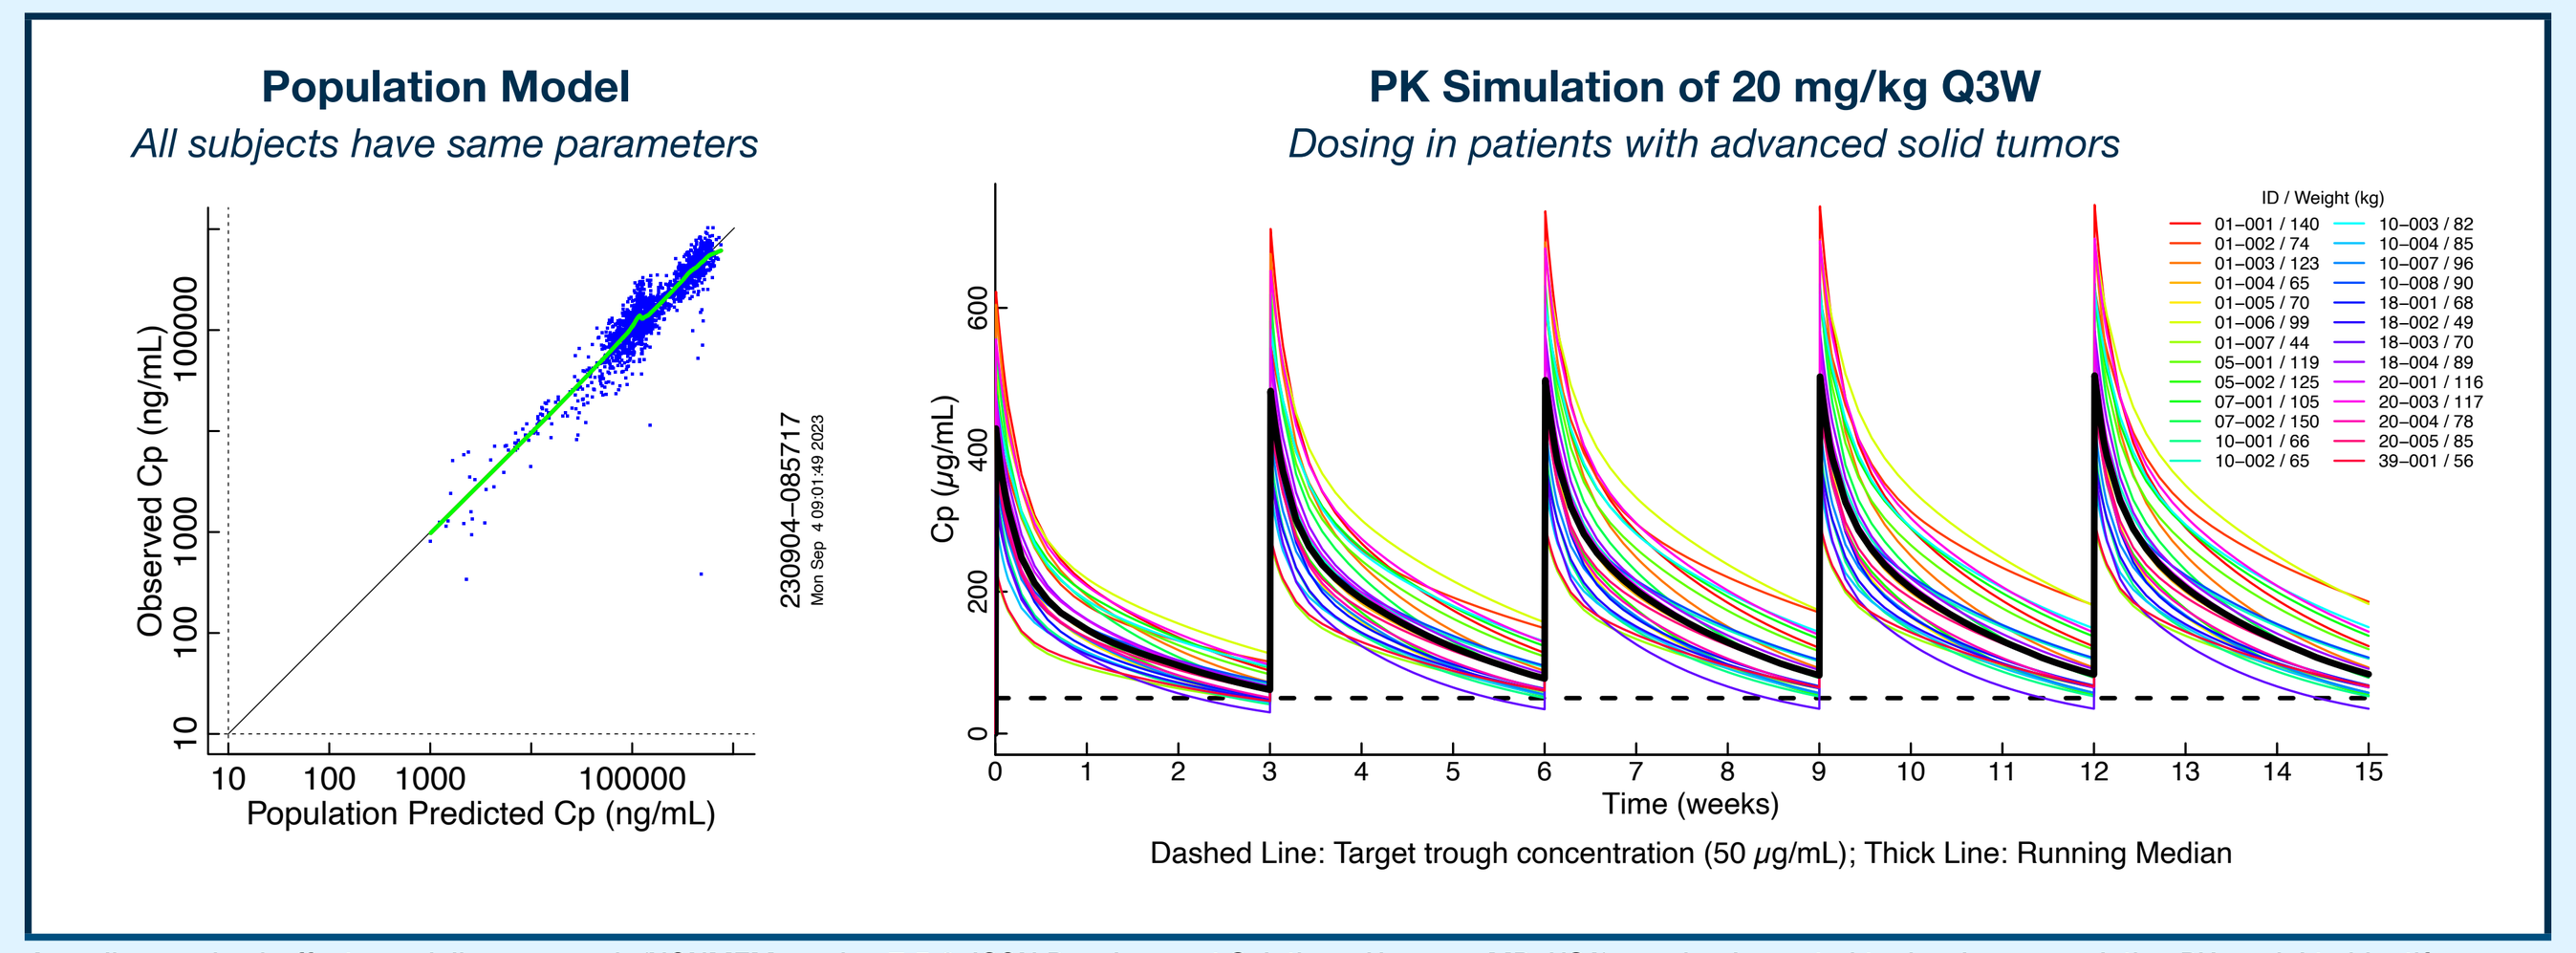
Population Model
All subjects have same parameters
Observed Cp (ng/mL)
Population Predicted Cp (ng/mL)
PK Simulation of 20 mg/kg Q3W
Dosing in patients with advanced solid tumors
Cp (μg/mL)
Time (weeks)
Dashed Line: Target trough concentration (50 μg/mL); Thick Line: Running Median

A nonlinear mixed-effects modeling approach (NONMEM version 7.5.0, ICON Development Solutions, Hanover, MD, USA) was implemented to develop a population PK model to identify potential covariates that correlate with drug clearance and to inform dosing decisions. Based on the simulations, the optimal biological dose was determined to be 20 mg/kg Q3W.

- Recommended dose for the expansion Phase 1b
 - Dose limiting toxicity not reached during the escalation of Phase 1a
 - Optimal biological dose of 20 mg/kg Q3W determined from minimum trough levels of 50 μg/ml, as anticipated from mechanistic modeling of receptor saturation
- Minimal evidence of TMDD; low levels of ADA detected in a limited number of patients
 - No evidence that post-dose positive titers associated with changes in clearance

Figure 5. TTX-080 Treatment Induces Gene Signature Changes in the Tumor Microenvironment

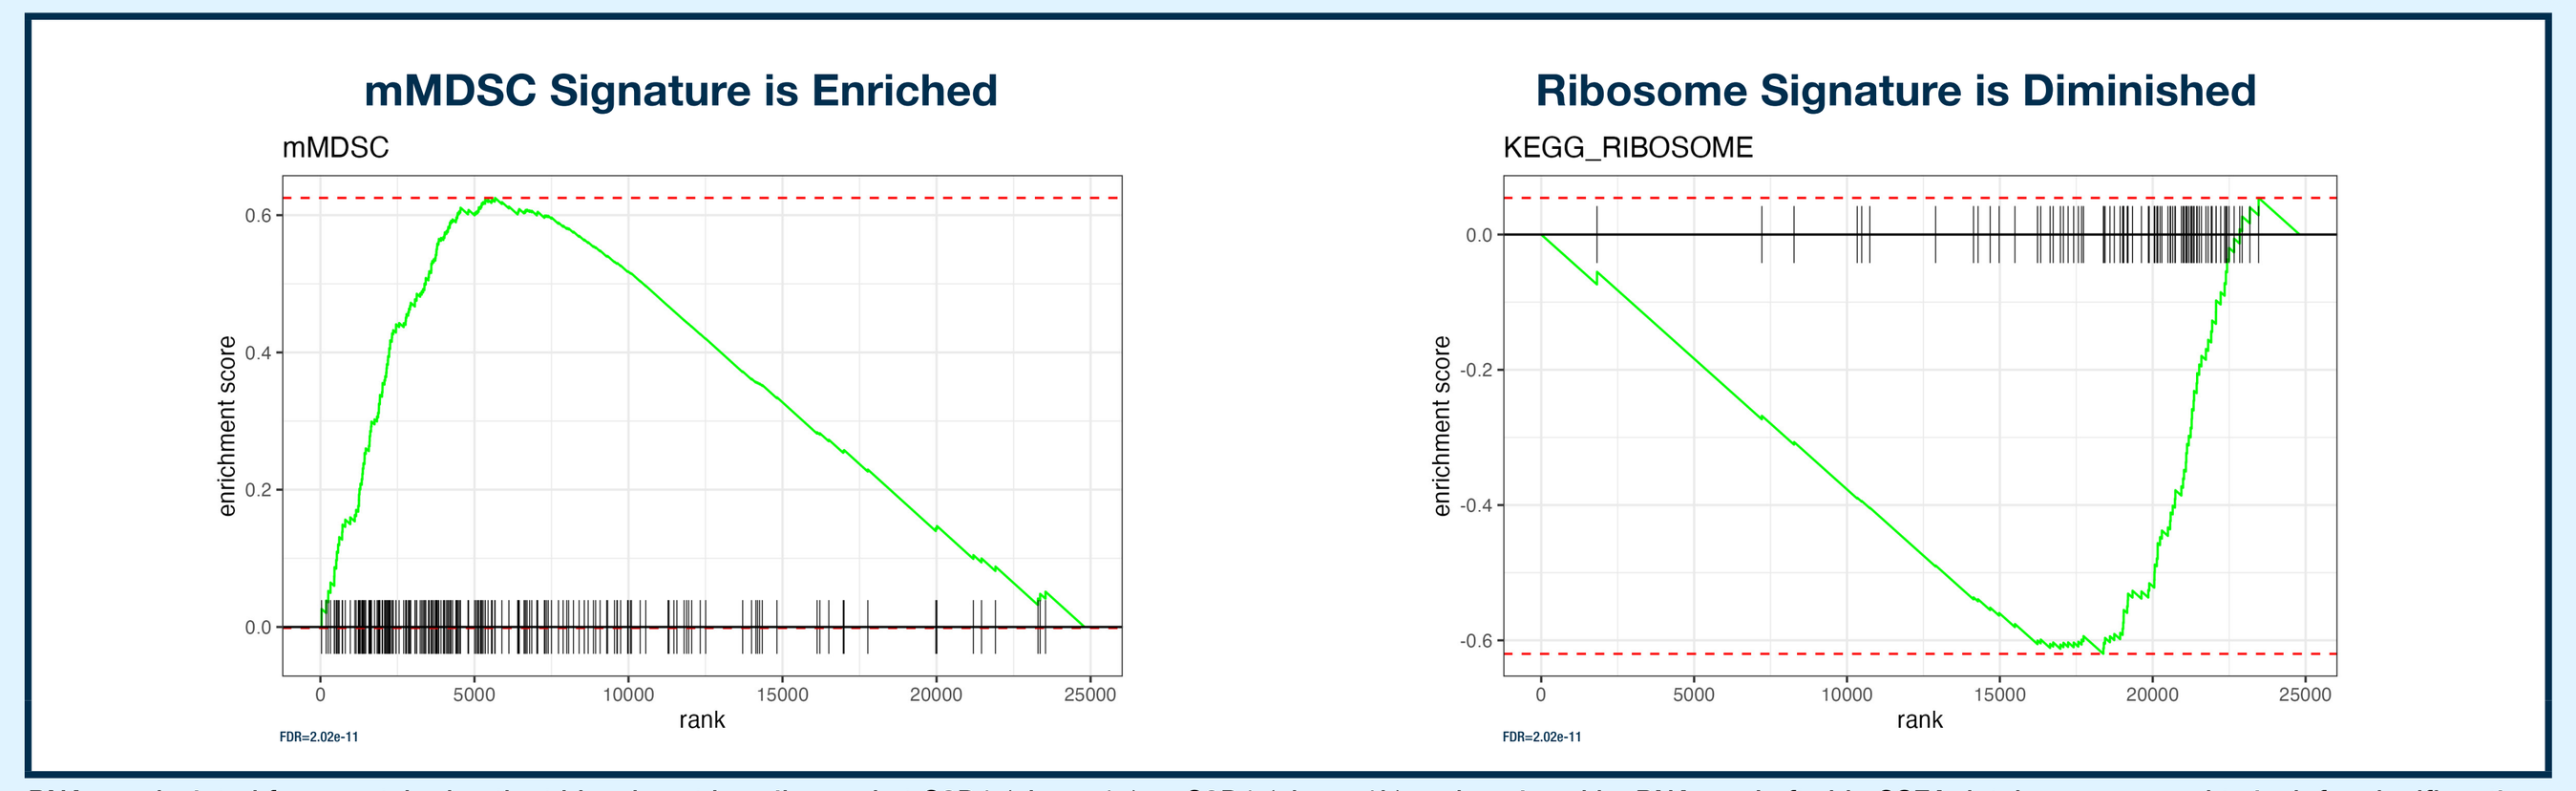
mMDSC Signature is Enriched
mMDSC
enrichment score
rank
Ribosome Signature is Diminished
KEGG_RIBOSOME
enrichment score
rank
FDR=2.02e-11

RNA was isolated from matched patient biopsies at baseline and at C2D1 (phase 1a) or C3D1 (phase 1b) and analyzed by RNAseq. Left side GSEA database was used to look for significantly upregulated gene sets post treatment. tz_mMDSC is enriched and KEGG_Ribosome is diminished, evaluated by CAMERA gene set enrichment post TTX-080 treatment. False Discovery Rate (FDR).

- Pre- versus post-treatment RNAseq isolated from tumor biopsies identified 97 gene sets with an FDR <.001
 - mMDSC signature is upregulated
 - Hypoxia genes are downregulated
 - Cancer signature gene sets are downregulated

Figure 6. TTX-080 Treatment Significantly Increases Immune-Related Gene Signatures in the Tumor

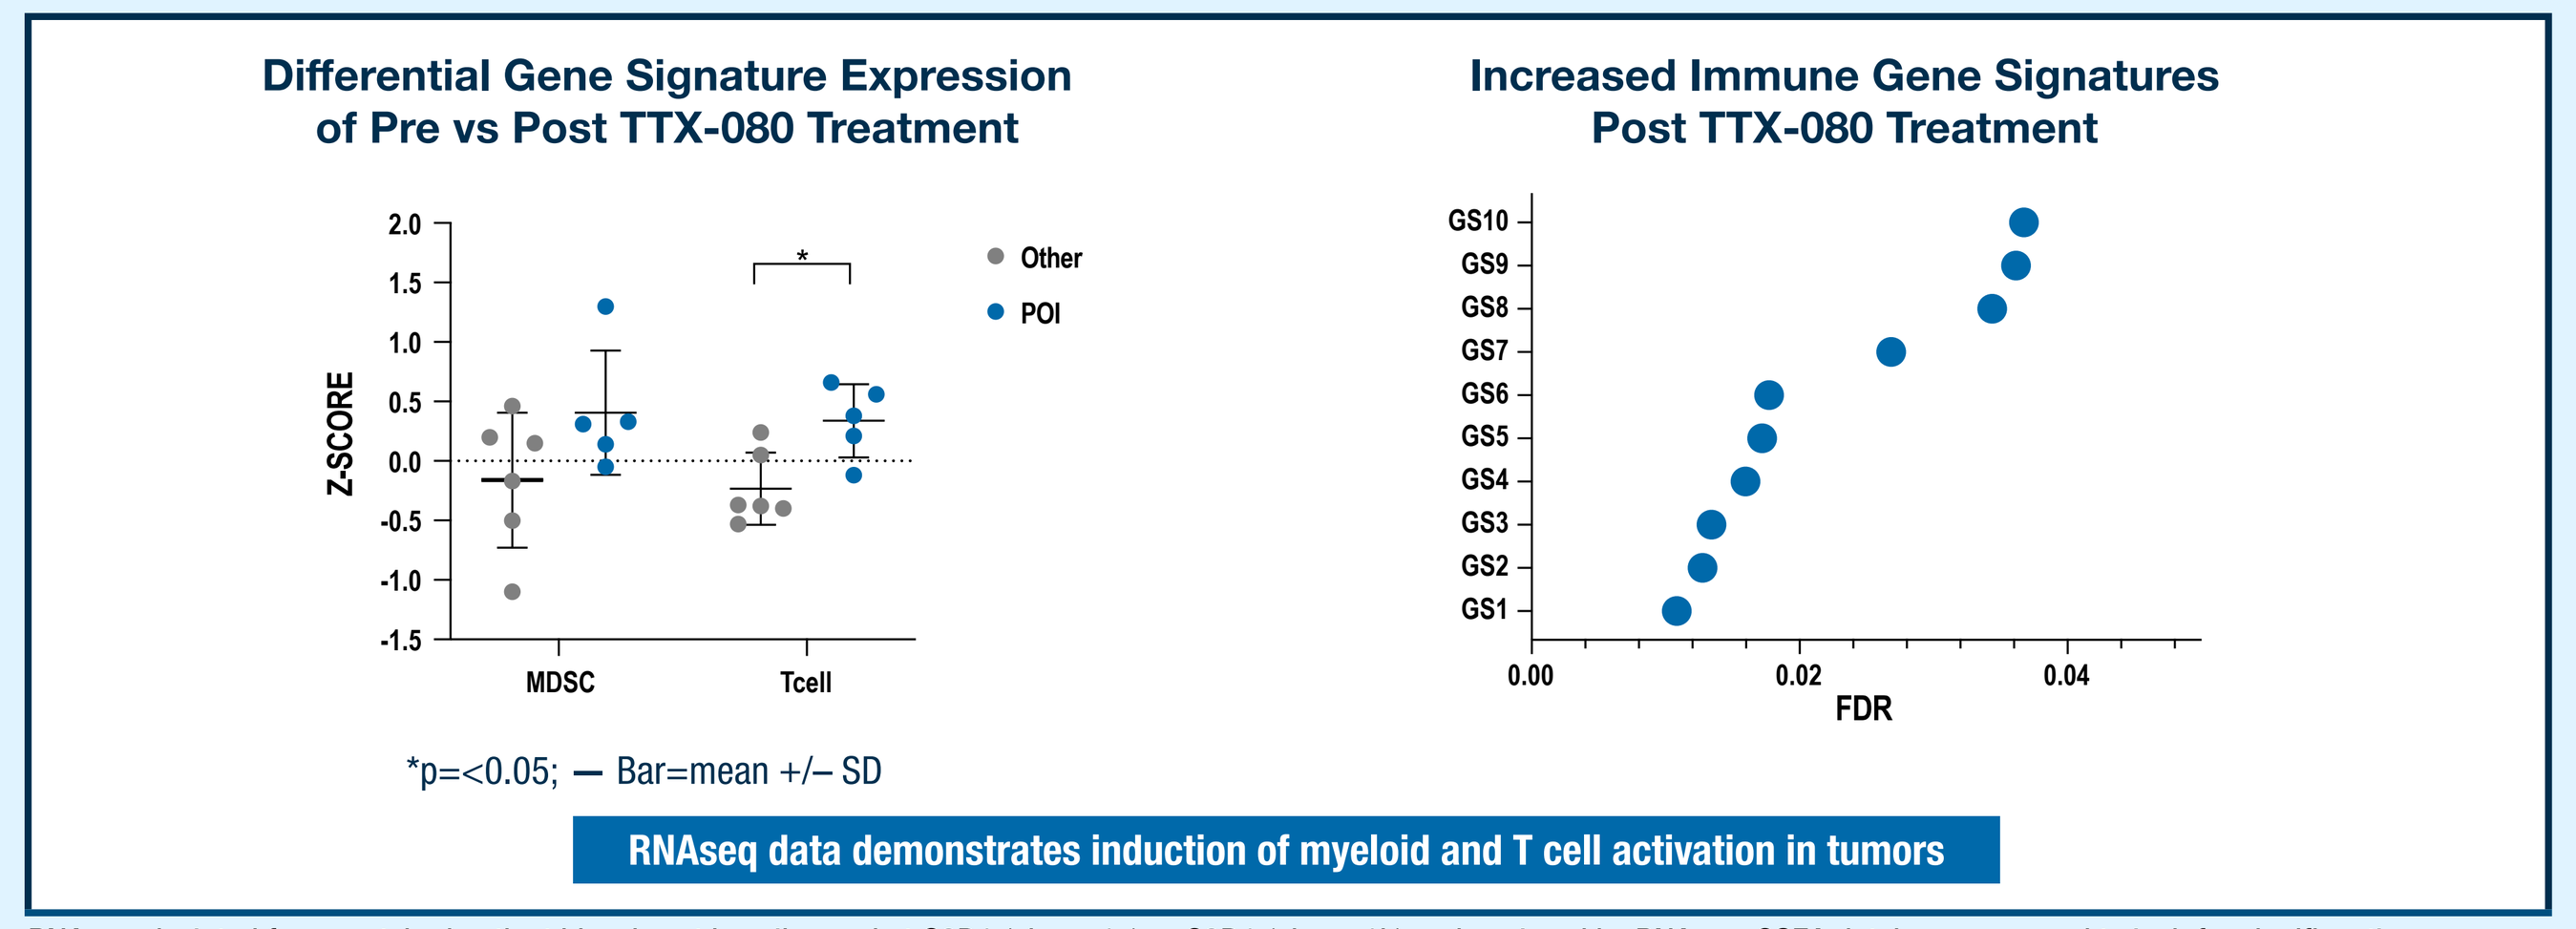
Differential Gene Signature Expression of Pre vs Post TTX-080 Treatment
Z-SCORE
MDSC
Tcell
Other
POI
*p=<0.05; — Bar=mean +/– SD
Increased Immune Gene Signatures Post TTX-080 Treatment
GS10
GS9
GS8
GS7
GS6
GS5
GS4
GS3
GS2
GS1
FDR
RNAseq data demonstrates induction of myeloid and T cell activation in tumors

RNA was isolated from matched patient biopsies at baseline and at C2D1 (phase 1a) or C3D1 (phase 1b) and analyzed by RNAseq. GSEA database was used to look for significantly upregulated gene sets post treatment evaluated by CAMERA gene set enrichment post TTX-080 treatment. False Discovery Rate (FDR).Patients tested either with TTX-080 monotherapy TTX-080 + cetuximab. Myeloid (MDSC) and Teff (Tcell) signatures are from Siu et al. *Clin Cancer Res.* 2022. Patients of interest include CR, PR, or SD >90 days.

Figure 7. TTX-080 Significantly Increased Myeloid Derived Chemokines CXCL9 and CXCL10

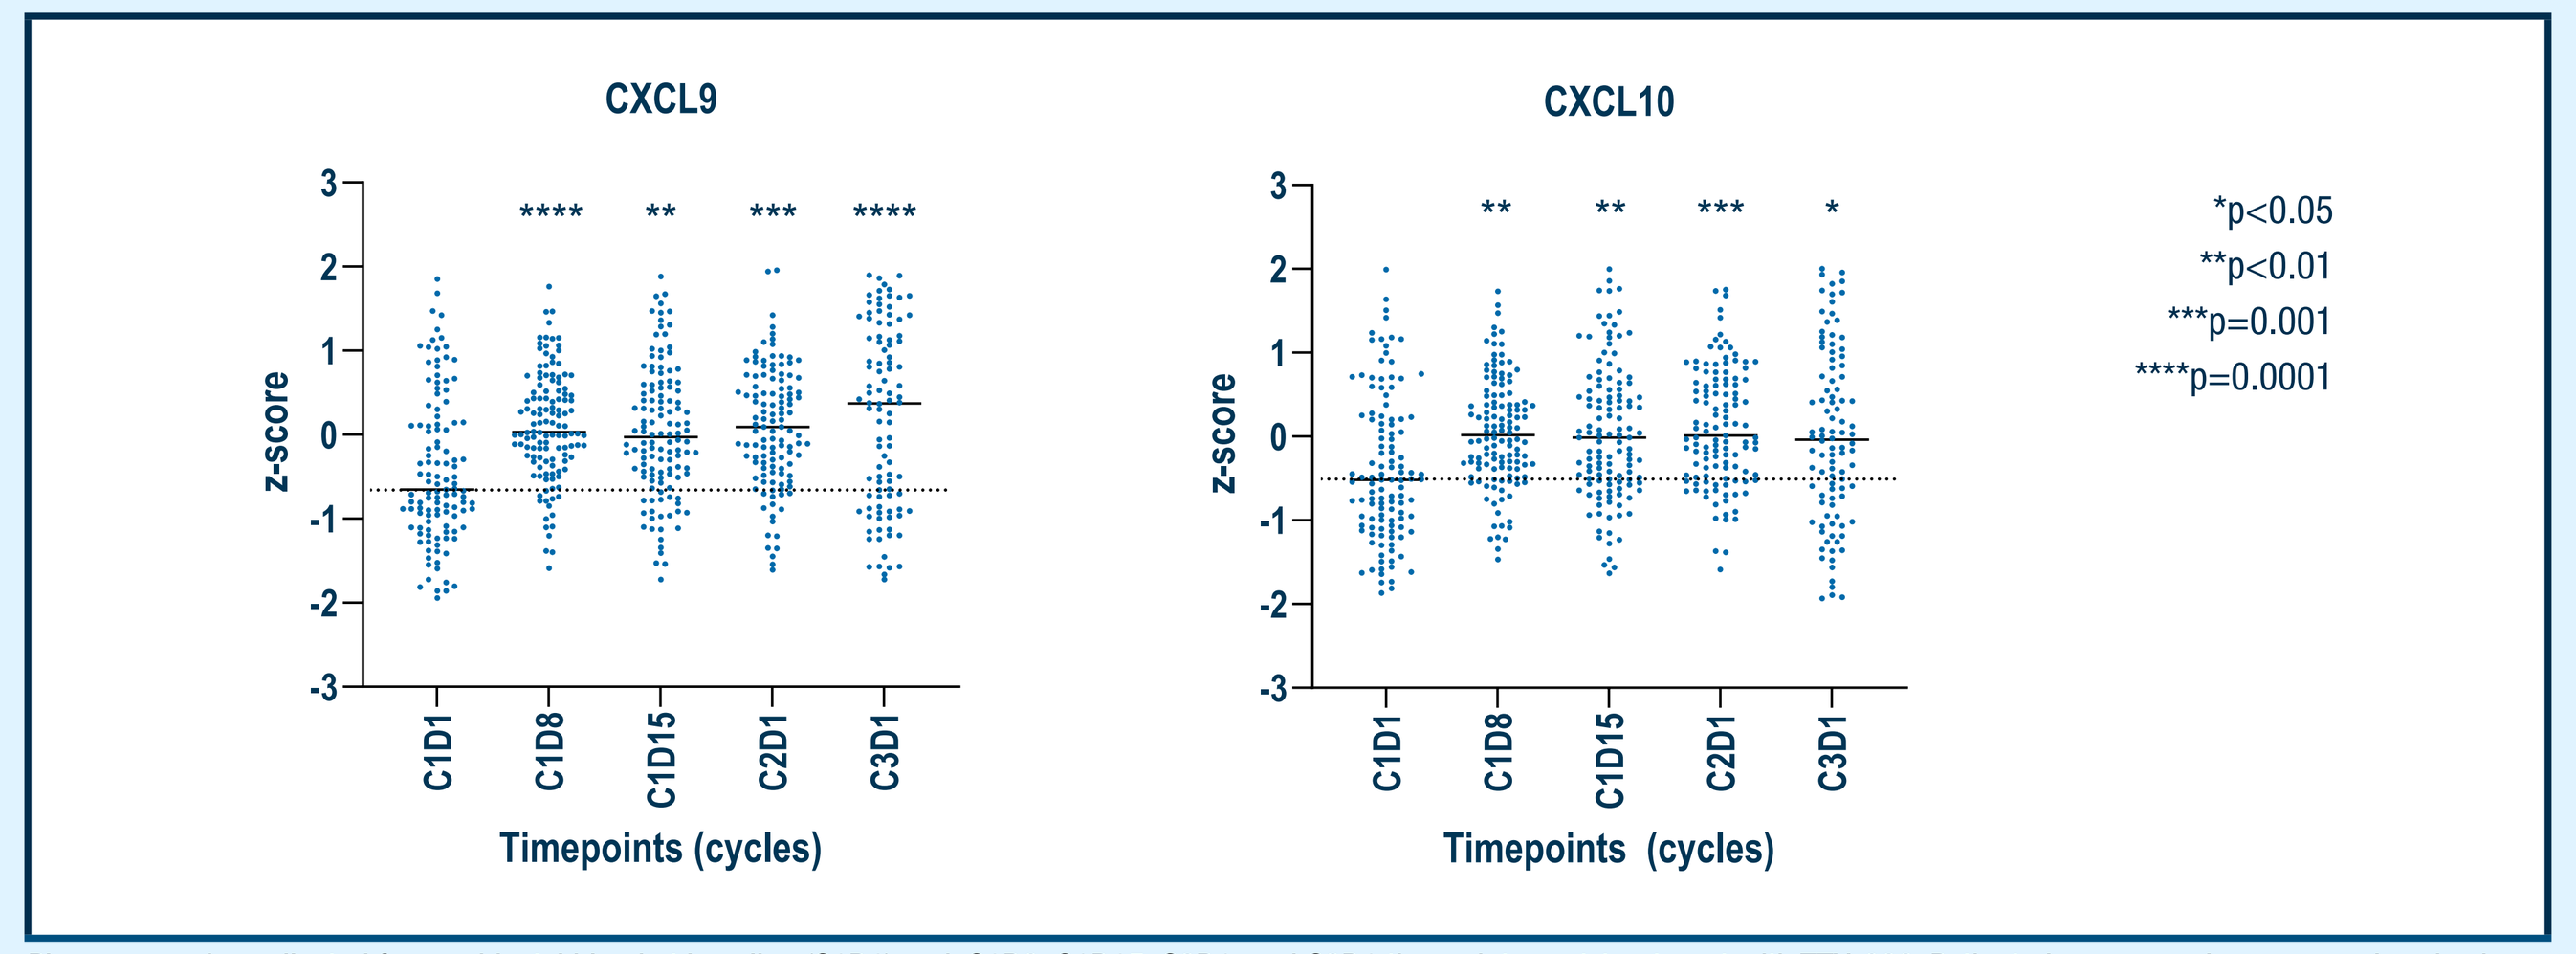
CXCL9
CXCL10
z-score
Timepoints (cycles)
C1D1
C1D8
C1D15
C2D1
C3D1
*p<0.05
**p<0.01
***p=0.001
****p=0.0001

Plasma samples collected from subjects' blood at baseline (C1D1) and, C1D8, C1D15, C2D1, and C3D1 timepoints post-treatment with TTX-080. Patient plasma samples were analyzed using the O-link® Target 96 Immuno-Oncology Panel for quantification of the chemokines. Z-SCORE: All samples tested, normalized by patient. P-values generated using Tukey-Kramer HSD.

Figure 8. Target-Related Changes in T Cell Populations Induced by TTX-080 – Unique to HLA-G/ILT2/ILT4 Pathway

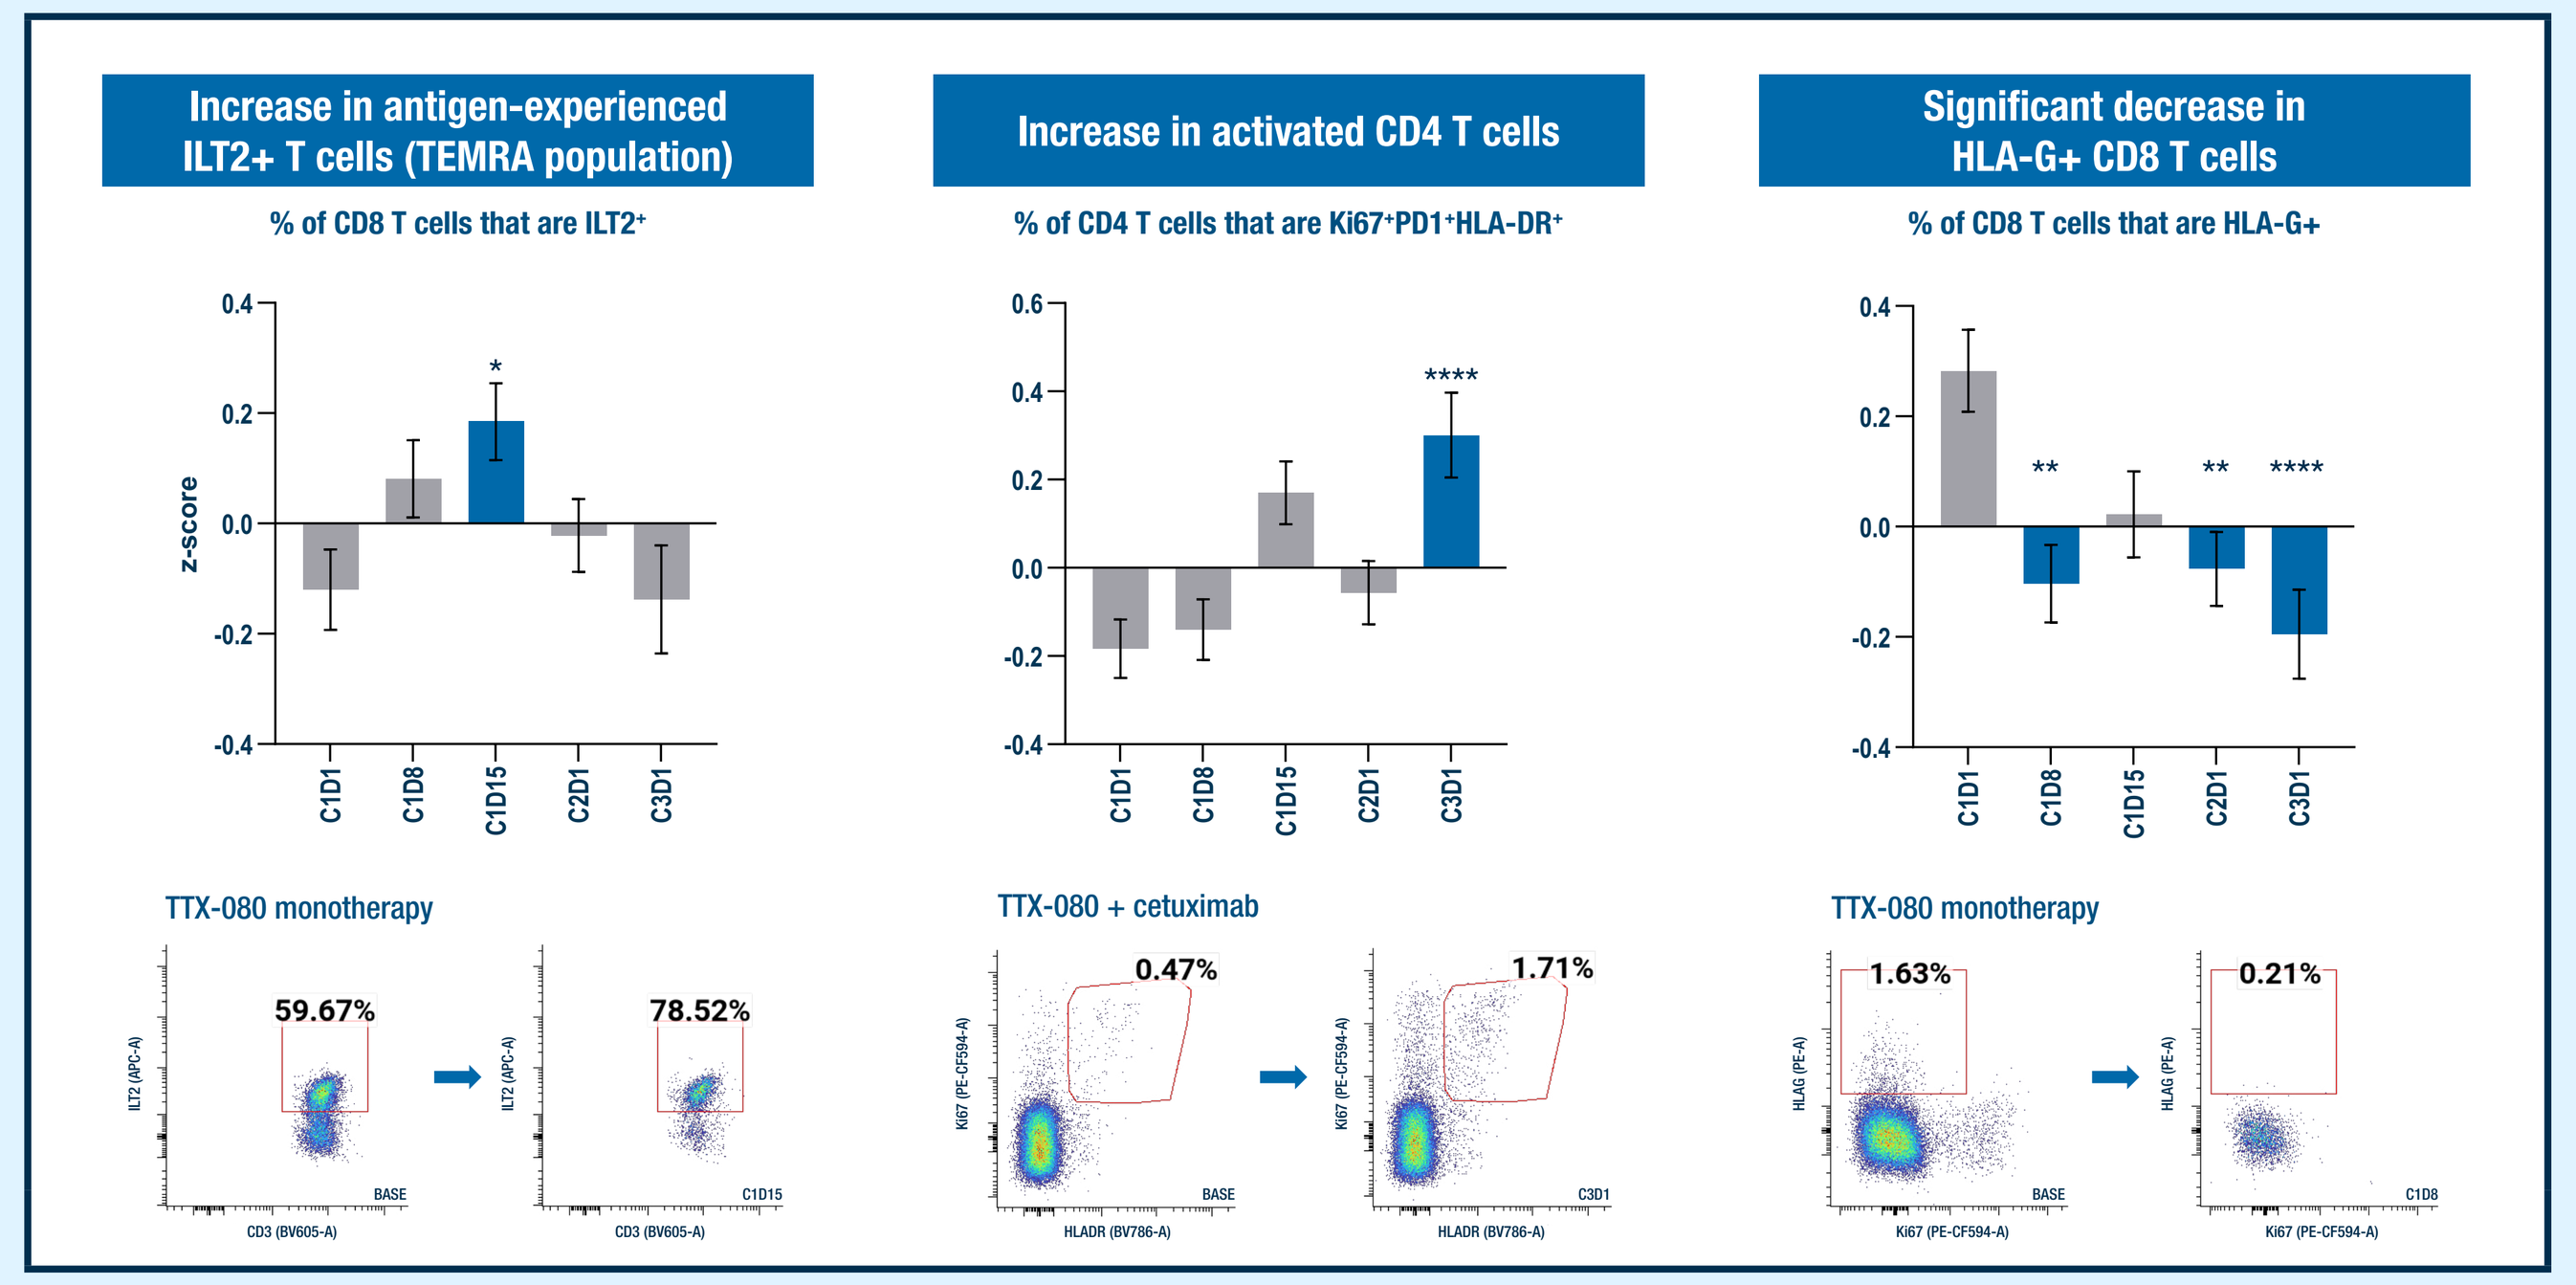
Increase in antigen-experienced ILT2+ T cells (TEMRA population)
% of CD8 T cells that are ILT2+
Increase in activated CD4 T cells
% of CD4 T cells that are Ki67+PD1+HLA-DR+
Significant decrease in HLA-G+ CD8 T cells
% of CD8 T cells that are HLA-G+
z-score
TTX-080 monotherapy
59.67%
78.52%
TTX-080 + cetuximab
0.47%
1.71%
TTX-080 monotherapy
1.63%
0.21%

Figure 9. TTX-080 Induced Innate Immunity by Increasing Activated NK cells

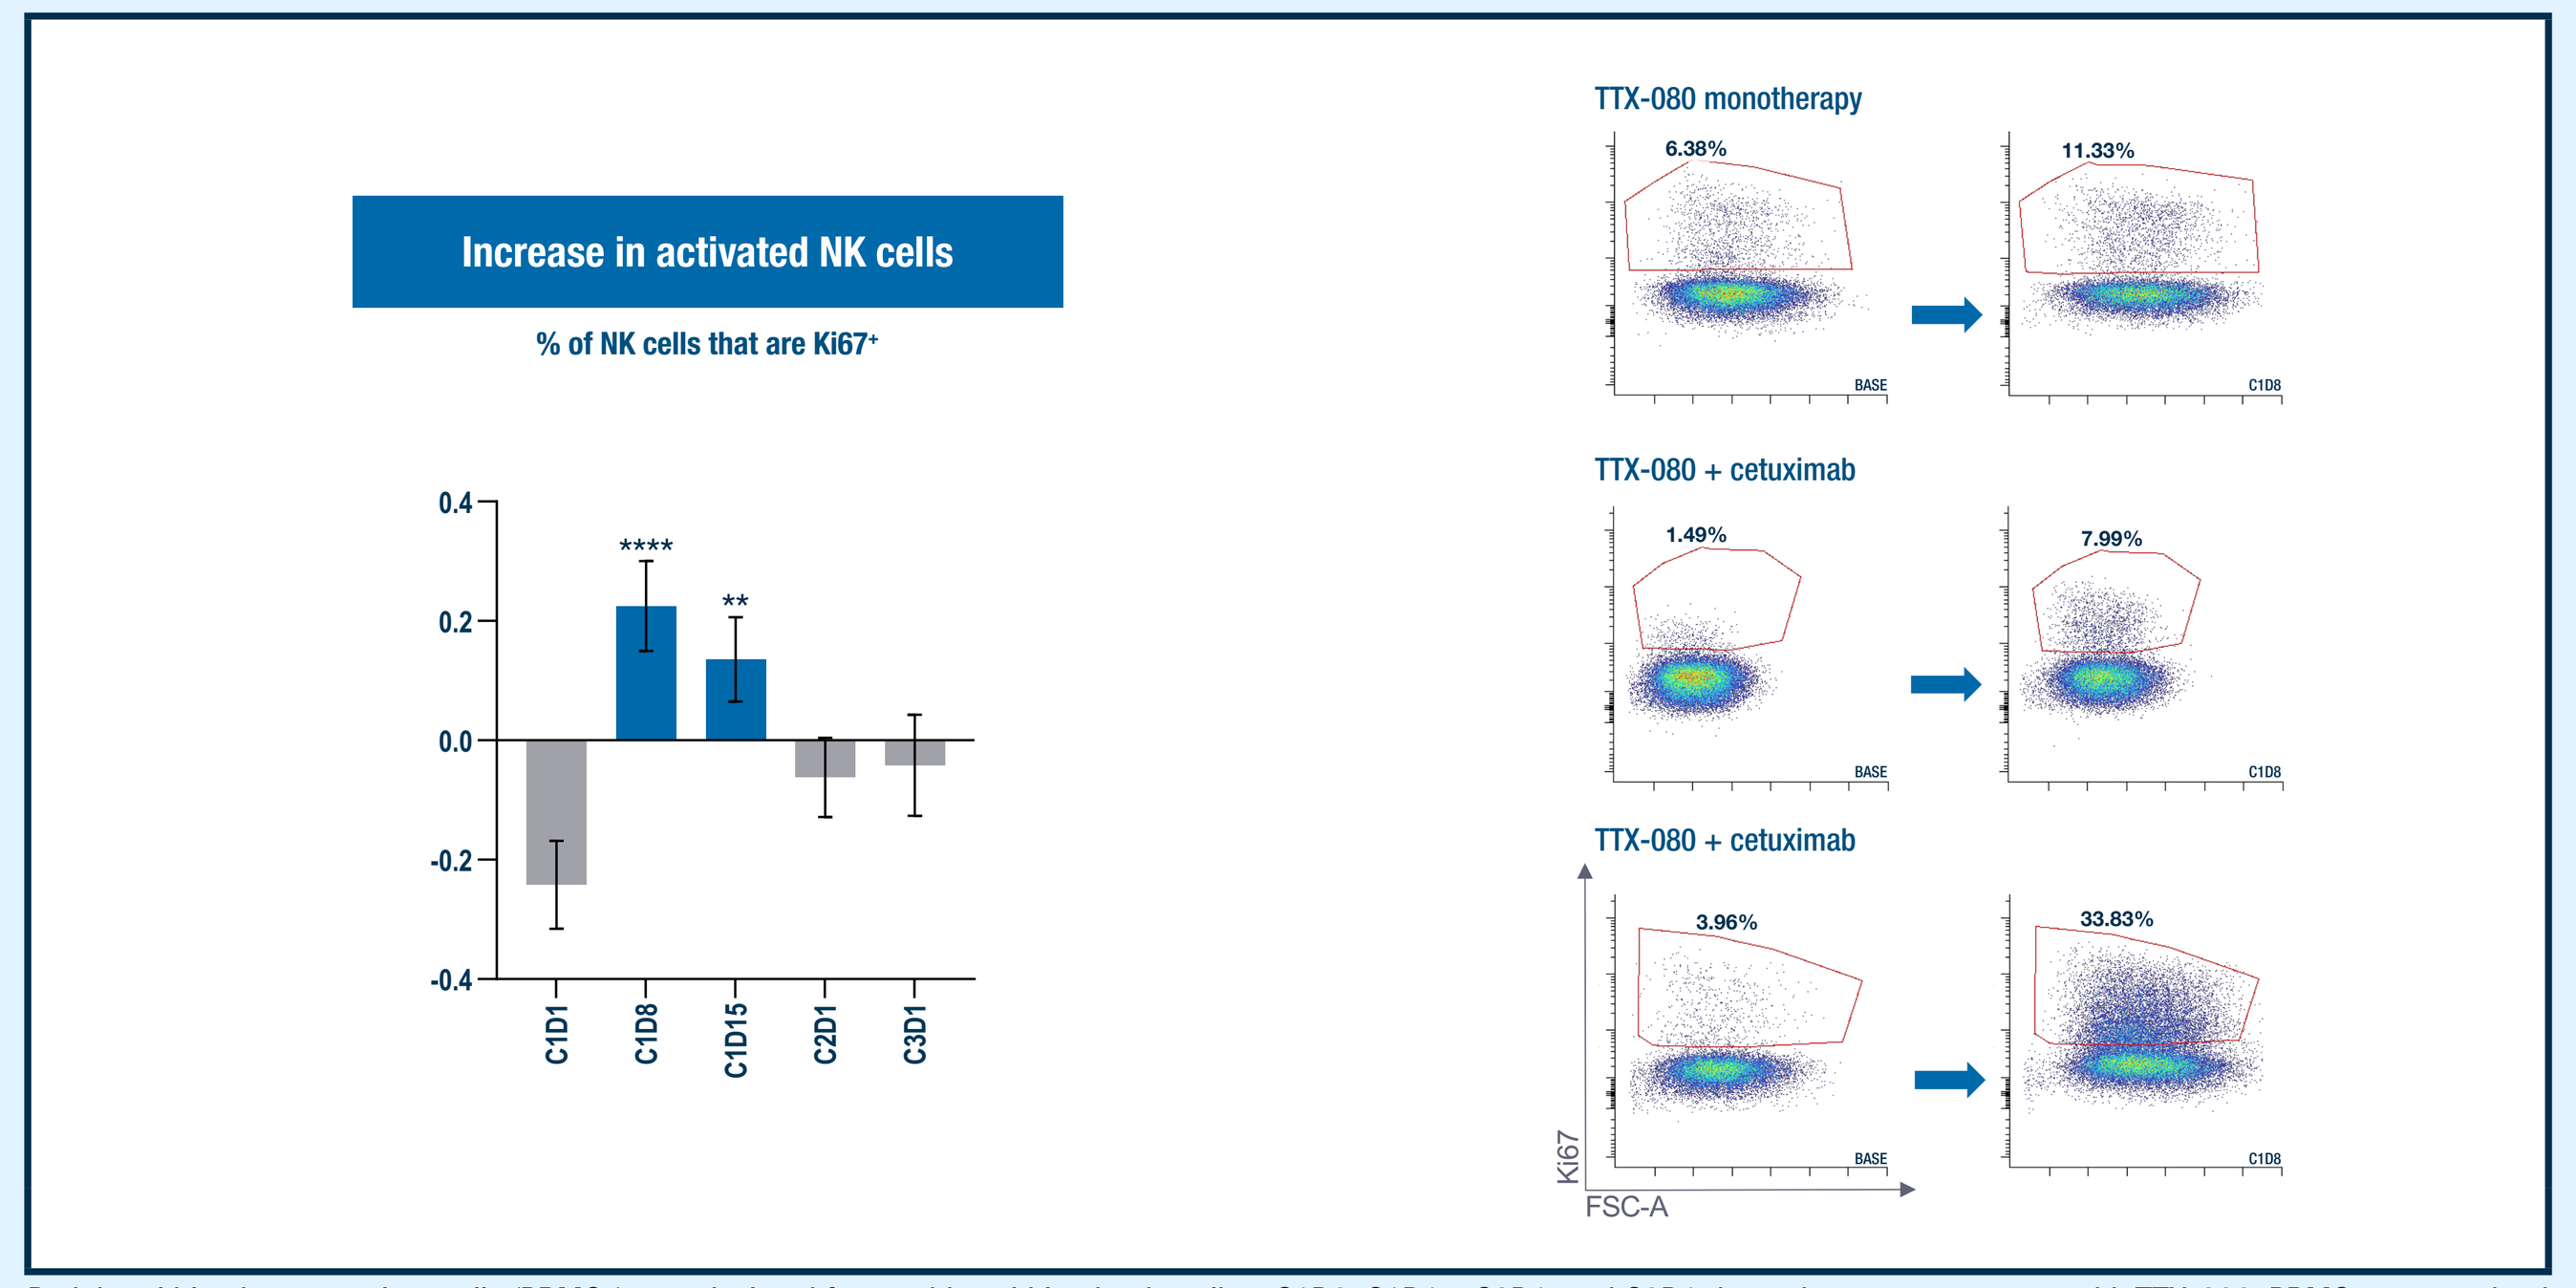
Increase in activated NK cells
% of NK cells that are Ki67+
TTX-080 monotherapy
6.38%
11.33%
TTX-080 + cetuximab
1.49%
7.99%
TTX-080 + cetuximab
3.96%
33.83%
Ki67
FSC-A

Peripheral blood mononuclear cells (PBMCs) were isolated from subjects' blood at baseline, C1D8, C1D15, C2D1, and C3D1 timepoints post-treatment with TTX-080. PBMCs were stained for cell surface and intracellular markers and detected by flow cytometry. Cell populations were analyzed via CellEngine® and statistical analysis using R. Statistical significance was determined using Tukey-Kramer HSD test.

Figure 10. Patients benefit from TTX-080 Treatment Regardless of TMB Status or PD-L1 Expression

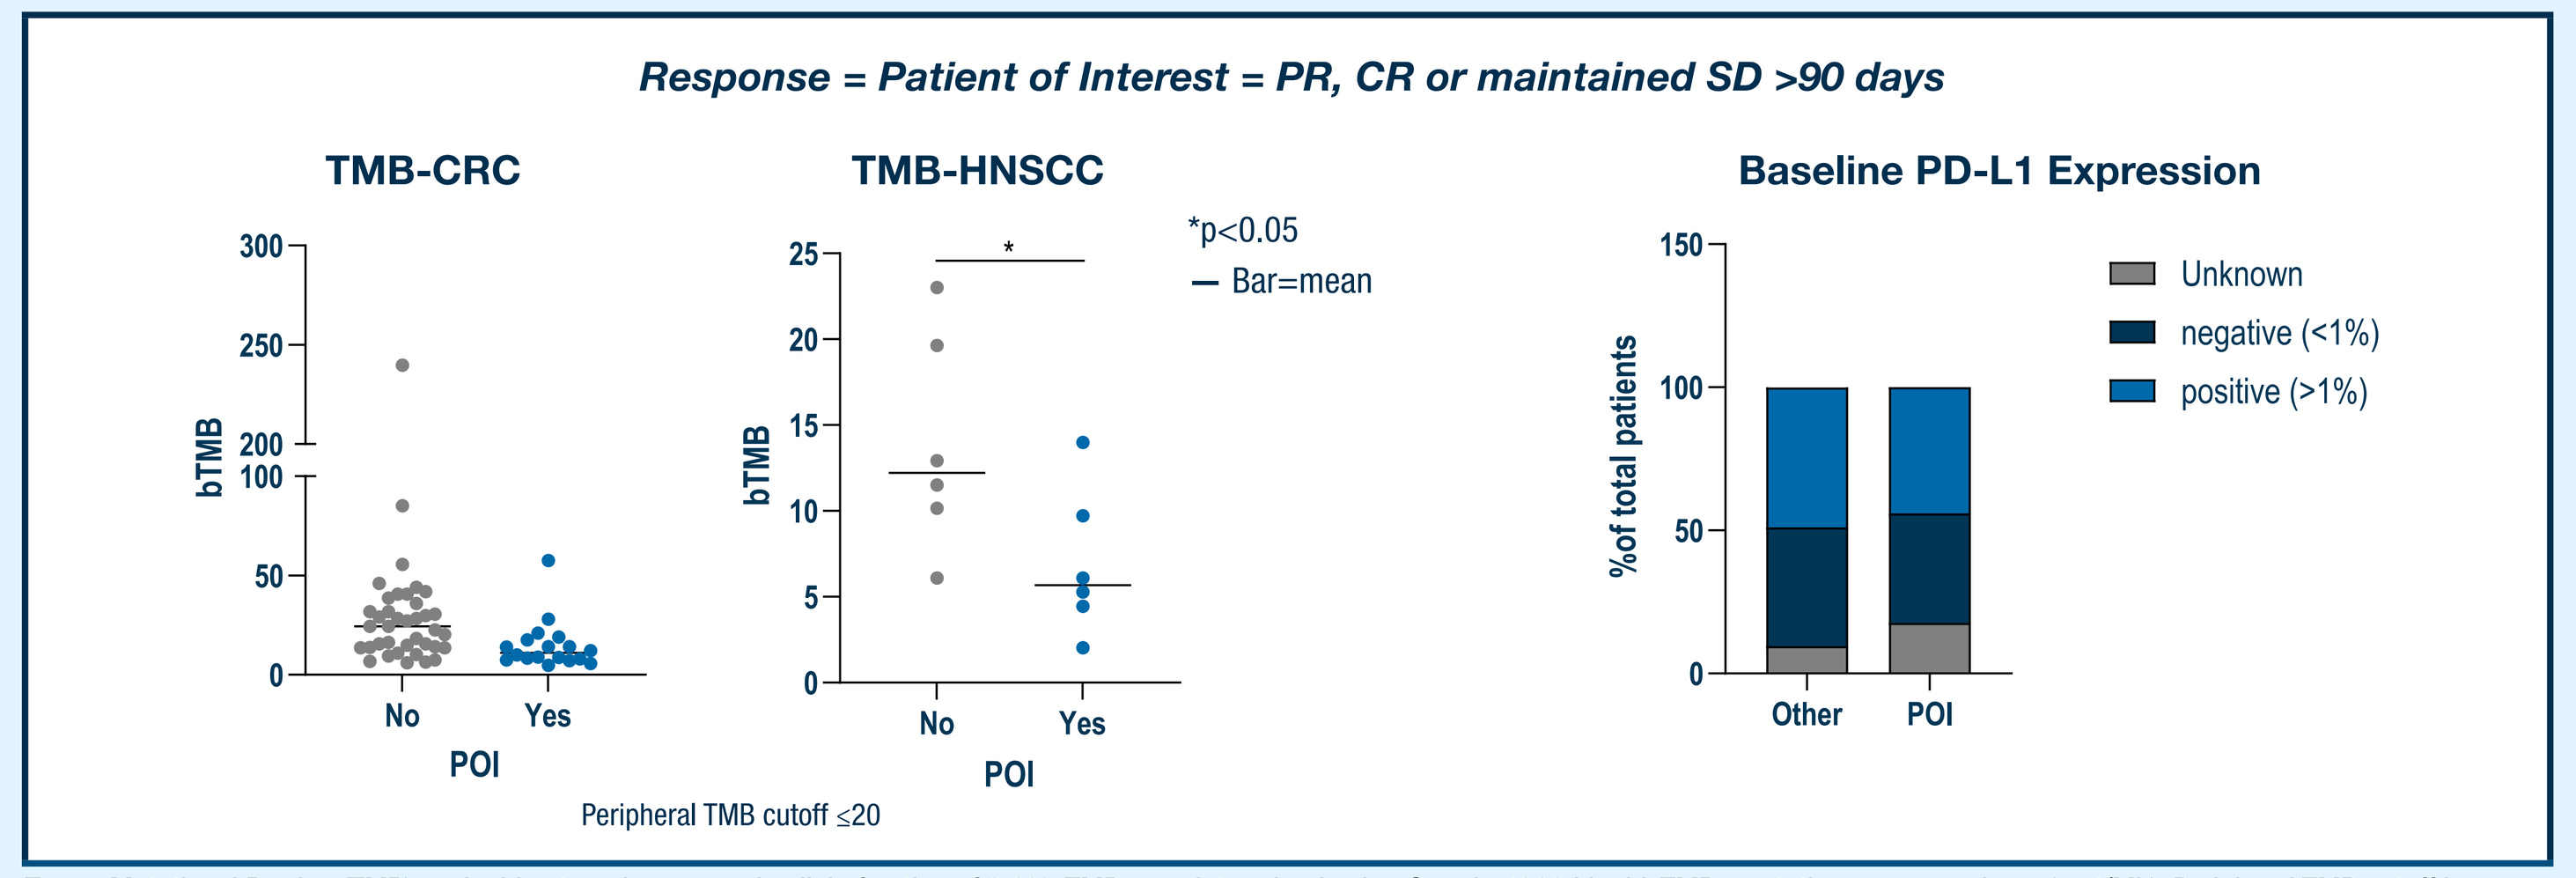
Response = Patient of Interest = PR, CR or maintained SD >90 days
TMB-CRC
TMB-HNSCC
bTMB
No
Yes
POI
*p<0.05
— Bar=mean
Peripheral TMB cutoff ≤20
Baseline PD-L1 Expression
%of total patients
Unknown
negative (<1%)
positive (>1%)
Other
POI

Tumor Mutational Burden (TMB) evaluable at or above sample allele fraction of 0.3%. TMB was determined using Guardant360 Liquid. TMB=mutations per megabase (mut/Mb). Peripheral TMB cutoff is 20 mut/Mb. Patients included in analysis are from mCRC and HNSCC cohorts combining TTX-080 + cetuximab. PD-L1 expression at baseline was either provided by investigators or determined by PD-L IHC PharmDX 22C3 CPS score. Statistical significance was determined using Tukey-Kramer HSD test.

Summary

TTX-080 Translational Summary

- Antibody exhibits drug-like properties supporting an optimal biological dose of 20 mg/kg Q3W
 - Minimal/no ADA
- Increases in the myeloid CXCL9 & CXCL10 chemokines are detected post-treatment
- Frequency changes in innate and adaptive immune cells are observed:
 - Increased ILT2+ CD8+ T cells (TEMRA – antigen experienced CD8+ T cells)
 - Ki67+PD1+HLA-DR+ CD4+ T cells (activation of adaptive immune cells)
 - Increased Ki67+ NK cells (innate immune activation)
 - Decreased HLA-G+ CD8+ T cells (on-mechanism)
- Treatment increases innate and adaptive immune gene signatures in the tumor
- Patients who benefit from treatment do not depend on TMB or PD-L1 status

Conclusions

TTX-080, A First-in-Pathway HLA-G Inhibitor with A Differentiated Mechanism of Action

- **Targets a novel checkpoint axis (HLA-G/ILT2/ILT4)** to restore both **innate & adaptive immune function—distinct from PD-1, PD-L1, & CTLA-4 inhibitors**
- **Translational analyses confirm on-mechanism immune activation,** supporting biological proof-of-concept in both blood & tumor
- **Preliminary clinical translational data** suggests that **mCRC and mHNSCC patients** treated with **TTX-080 in combination with cetuximab** may benefit from treatment **regardless of TMB or PD-L1 status**
- **Ongoing randomized study** of **TTX-080 + cetuximab + FOLFIRI** in **metastatic colorectal cancer (NCT04485013)**

Gene Signature #	Gene Signature Name	Gene Signature #	Gene Signature Name
GS1	GSE45365_WT_VS_IFNAR_KO_CD11B_DC_MCMV_INFECTION_DN	GS6	GSE41867_NAIVE_VS_DAY15_LCMV_CONE13_EFFECTOR_CD8_TCELL_UP
GS2	GSE41867_DAY6_VS_DAY15_LCMV_CLONE13_EFFECTOR_CD8_TCELL_DN	GS7	GSE12484_HEALTHY_VS_PERIDONTITIS_NEUTROPHILS_UP
GS3	HOWARD_NK_CELL_INACT_MONOV_INFLUENZA_A_INDONESIA_05_2005_H5N1_AGE_18_49YO_3DY_UP	GS8	GSE18281_CORTICAL_THYMOCYTE_VS_WHOLE_CORTEX_THYMUS_UP
GS4	GSE41176_UNSTIM_VS_ANTI_IGM_STIM_TAK1_KO_BCELL_3H_DN	GS9	GSE21379_TFH_VS_NON_TFH_CD4_TCELL_DN
GS5	GSE37605_C57BL6_VS_NOD_FOXP3_FUSION_GFP_TREG_UP	GS10	GSE37532_VISCERAL_ADIPOSE_TISSUE_VS_LN_DERIVED_TCONV_CD4_TCELL_DN

Abbreviations

ADA, anti-drug antibodies; AML, acute myeloid leukemia; CRC, colorectal cancer; C[X]D[X], Cycle X, Day X; FDR, false discovery rate; HNSCC, head and neck squamous cell carcinoma; HSD, honestly significant difference; MHC, major histocompatibility complex; mMDSC, monocytic myeloid-derived suppressor cells; MSS, microsatellite stable; NK, natural killer; NONMEM, nonlinear mixed-effects model; NSCLC, non-small cell lung cancer; PK, pharmacokinetics; POC, proof of concept; RCC, renal cell carcinoma; SCC, squamous cell carcinoma; TEMRA, terminally differentiated effector memory T cells re-expressing CD45RA; TMB, tumor mutational burden; TMDD, target-mediated drug disposition; TNBC, triple negative breast cancer; tz_mMDSC, Tizona-defined monocytic myeloid-derived suppressor cells.

Acknowledgements

Thank you to the patients who participated in this clinical study.
Thank you to Dennig Marketing Group for assistance in assembling this poster.

Society for Immunotherapy of Cancer (SITC), November 5-9, 2025, National Harbor, MD

Email: cbeers@tizonatx.com